Abstract number 291

Leiden University Medical Center

Mark J.A. Schoonderwoerd[1], Madelon Paauwe[1], Ricardo Angela[1], Maaike Koops[1], Tom Harrijvan[1], Cornelis F.M. Sier[2], Marieke Fransen[3], Charles P. Theuer[4], Lukas J.A.C. Hawinkels[11]
Departments of [1]Gastroenterology-Hepatology, [2]Surgery, [3]Immunohematology and Blood Transfusion, Leiden University Medical Center, Leiden, The Netherlands, [4]Tracon Pharmaceuticals, San Diego, CA, USA,

Synergistic inhibition of cancer invasion and metastasis by combined anti-PD1/TRC105-mediated Endoglin targeting on cancer-associated fibroblasts and endothelial cells

Introduction

Endoglin is a co-receptor for Transforming Growth Factor (TGF)-β and Bone Morphogenetic Protein (BMP)-9 and has a crucial role in developmental and tumor angiogenesis.

Endoglin is highly expressed by angiogenic endothelial cells, but also by cancer-associated fibroblasts (CAFs) at the invasive margins of colorectal tumors (Paauwe *et al* Clin Ca Res 2018) and several other cells in the tumor microenvironment. Expression of endoglin has been correlated with metastases and poor prognosis in cancer patients.

TRC105 (carotuximab, Tracon pharmaceuticals) is a humanized IgG1 antibody directed against endoglin and was previously shown to inhibit tumor angiogenesis *in vitro* and *in vivo*. The effects of TRC105 seem to be dependent on an immune response against the target cells. TRC105 is currently in clinical development in multiple phase 2 and -3 clinical trials as well as a phase 1 trial with nivolumab.

In this project we have evaluated if immune responses against endoglin can be enhanced by simultaneously activating anti-tumor responses using anti-PD1 therapy. We used three different mouse colorectal cancer (CRC) models to evaluate therapeutic responses.

Aim:

1. To evaluate if combining TRC105 with anti-PD1 enhances anti-tumor responses in three mouse models for different stages of colorectal cancer.

2. To evaluate the underlying mechanism of action of synergistic effects.

Materials and methods

- Chemically induced CRC model, using azoxymethane (AOM) followed by dextran sodium sulphate (DSS) induced inflammation
- Syngeneic orthotopic or subcutaneous tumor models using MC38 or CT26 mouse CRC cells
- Upon tumor formation (palpable tumors) treatment with TRC105 and/or anti-PD1 therapy (or isotype control antibodies)
- Endpoint: tumor > 1,5 cm^3 (survival) or short term (9 days)
- Flow cytometry analysis, immunohistochemical analysis

Results

Tumor formation was induced by injection with AOM, followed by 3 cycles of DSS. After 84 days mice were sacrificed and number of lesions was evaluated.

Azoxymethane-DSS model (colitis-associated cancer)

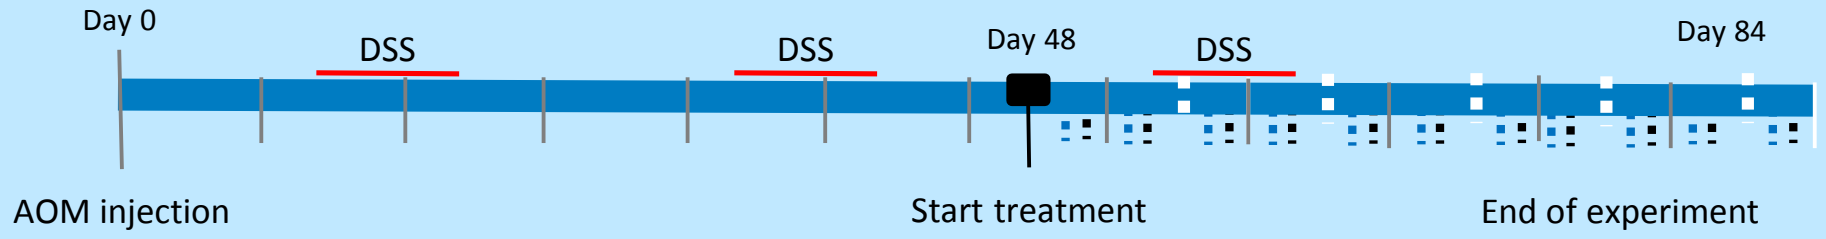
Day 0
DSS
DSS
Day 48
DSS
Day 84
AOM injection
Start treatment
End of experiment

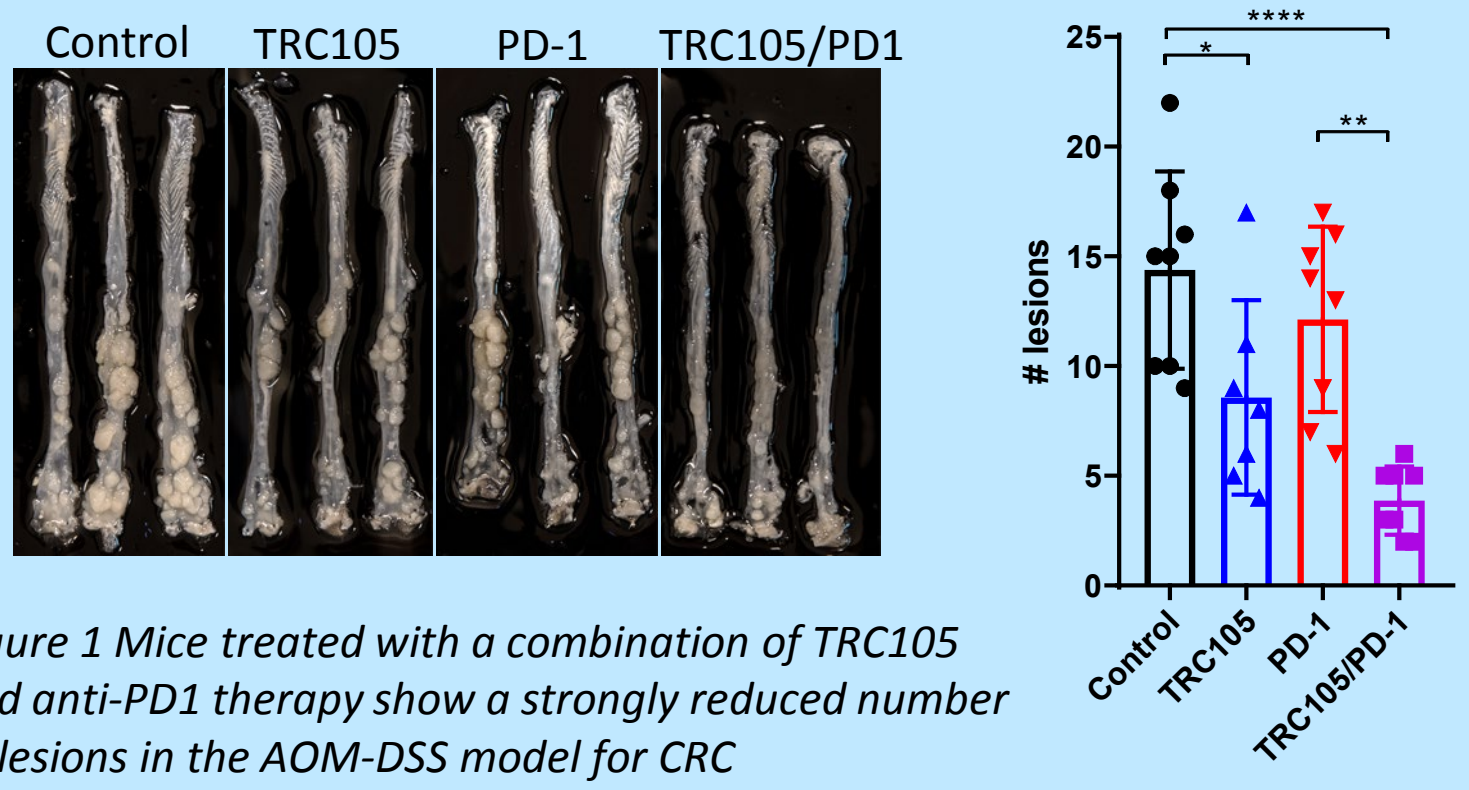
Control
TRC105
PD-1
TRC105/PD1
lesions
Control
TRC105
PD-1
TRC105/PD-1

Figure 1 Mice treated with a combination of TRC105 and anti-PD1 therapy show a strongly reduced number of lesions in the AOM-DSS model for CRC

Orthotopic MC38 CRC model (late stage/metastatic cancer)

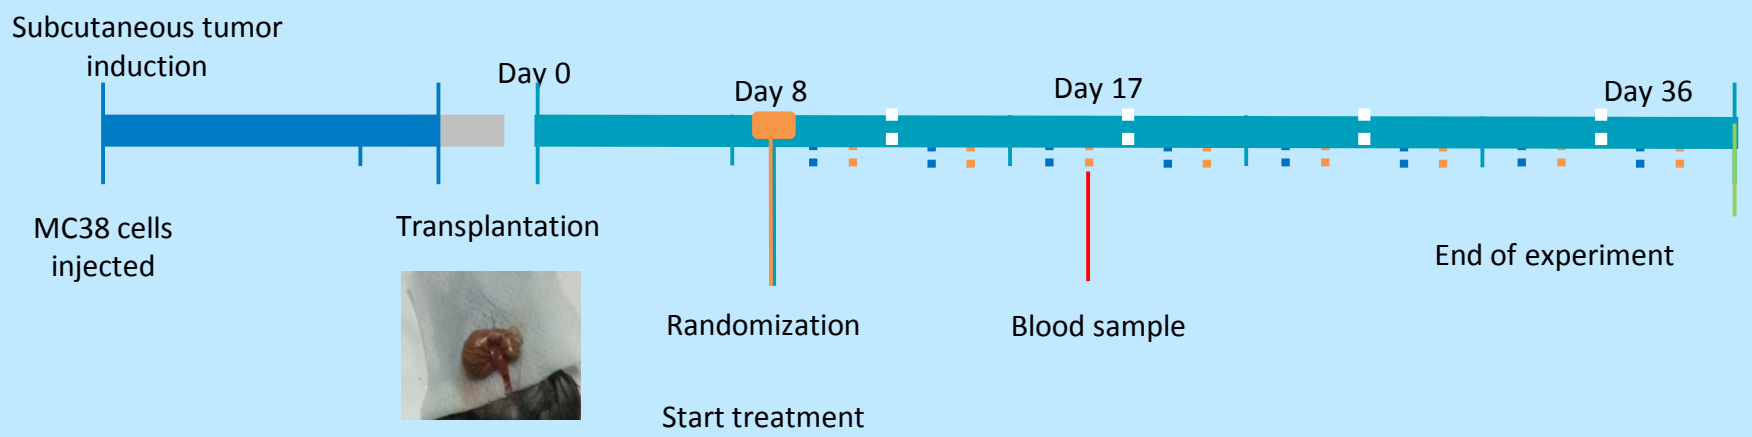
Subcutaneous tumor induction
Day 0
Day 8
Day 17
Day 36
MC38 cells injected
Transplantation
Randomization
Start treatment
Blood sample
End of experiment

After tumor transplantation mice were divided in groups based on equal bioluminescence signal and treated with antibodies. After 36 days mice were sacrificed and tumor volume (A), vascularization (B) and the number of tumor-free animals was determined.

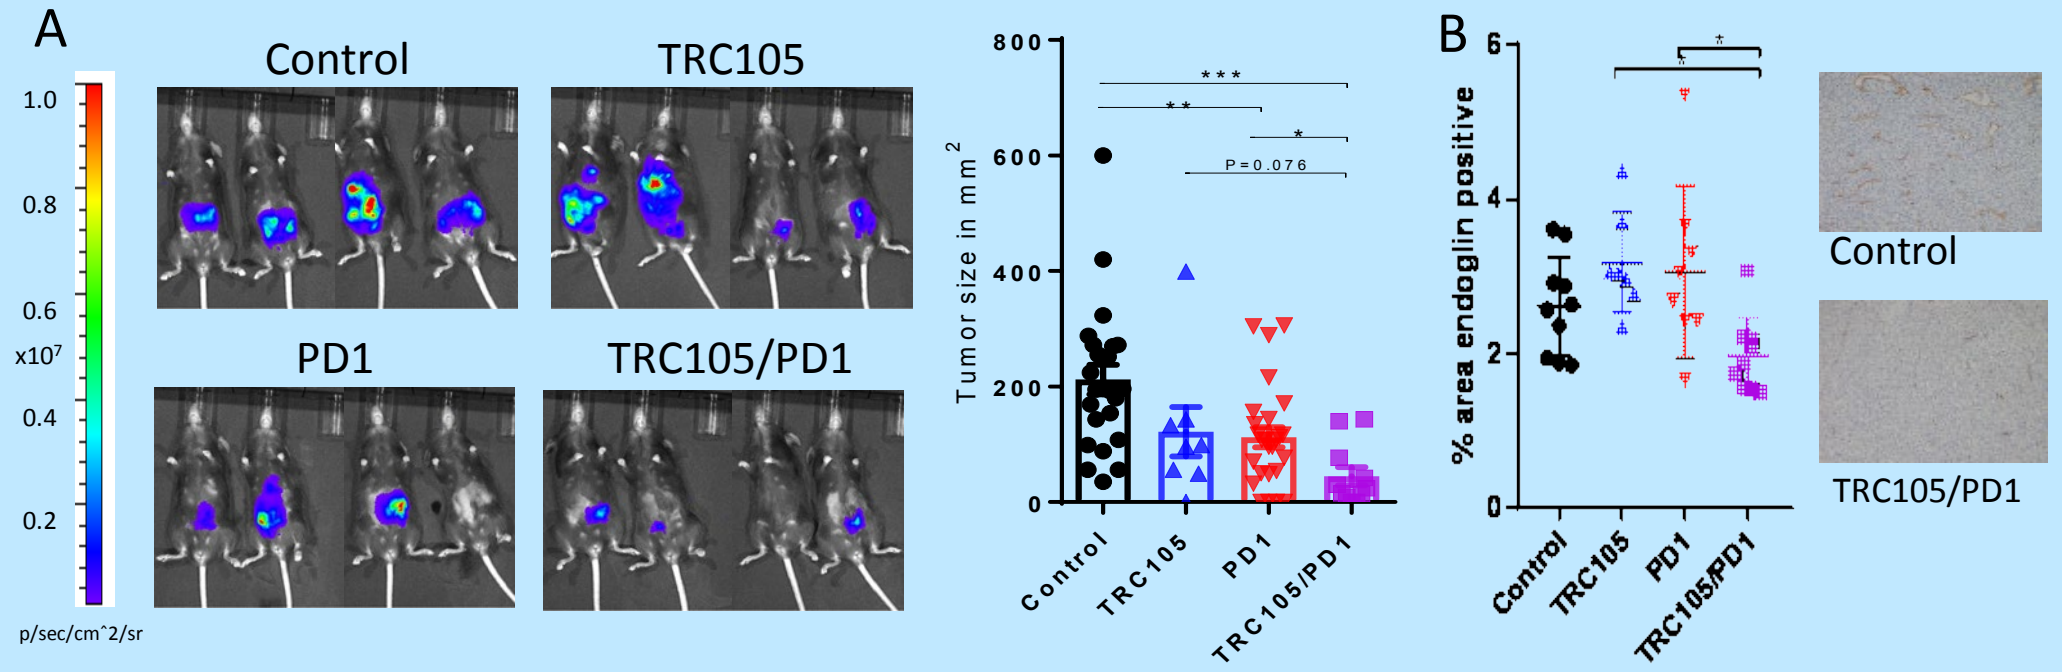
A
Control
TRC105
PD1
TRC105/PD1
p/sec/cm^2/sr
Tumor size in mm2
P=0.076
Control
TRC105
PD1
TRC105/PD1
B
% area endoglin positive
Control
TRC105
PD1
TRC105/PD1
Control
TRC105/PD1

Figure 2 Mice treated with a combination of TRC105 and anti-PD1 therapy display significantly reduced tumor volume. An increased number of tumor-free mice is detected.

MC38/CT26 subcutaneous tumor model

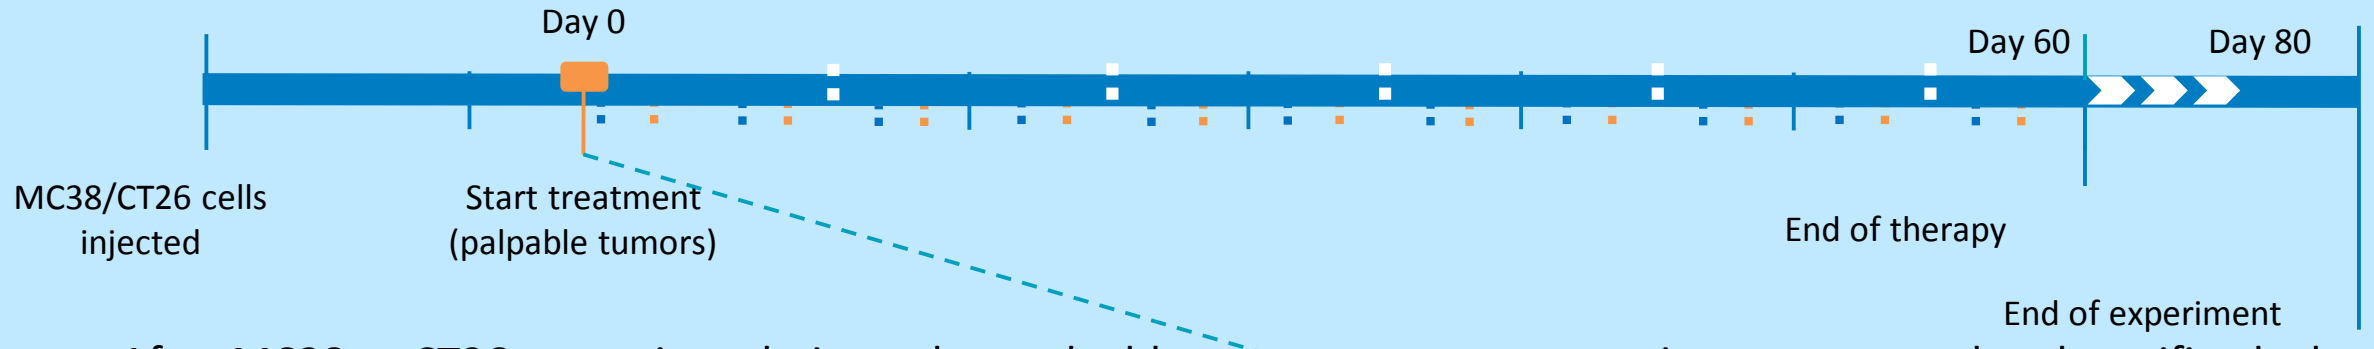
Day 0
Day 60
Day 80
MC38/CT26 cells injected
Start treatment (palpable tumors)
End of therapy
End of experiment

After MC38 or CT26 tumor inoculation, when palpable tumors were present mice were treated and sacrificed when tumors reached 1.5 cm2. In a second experiment (Fig 3E) the immune infiltrate was analyzed 9 days after start of treatment, when tumors had equal volume.

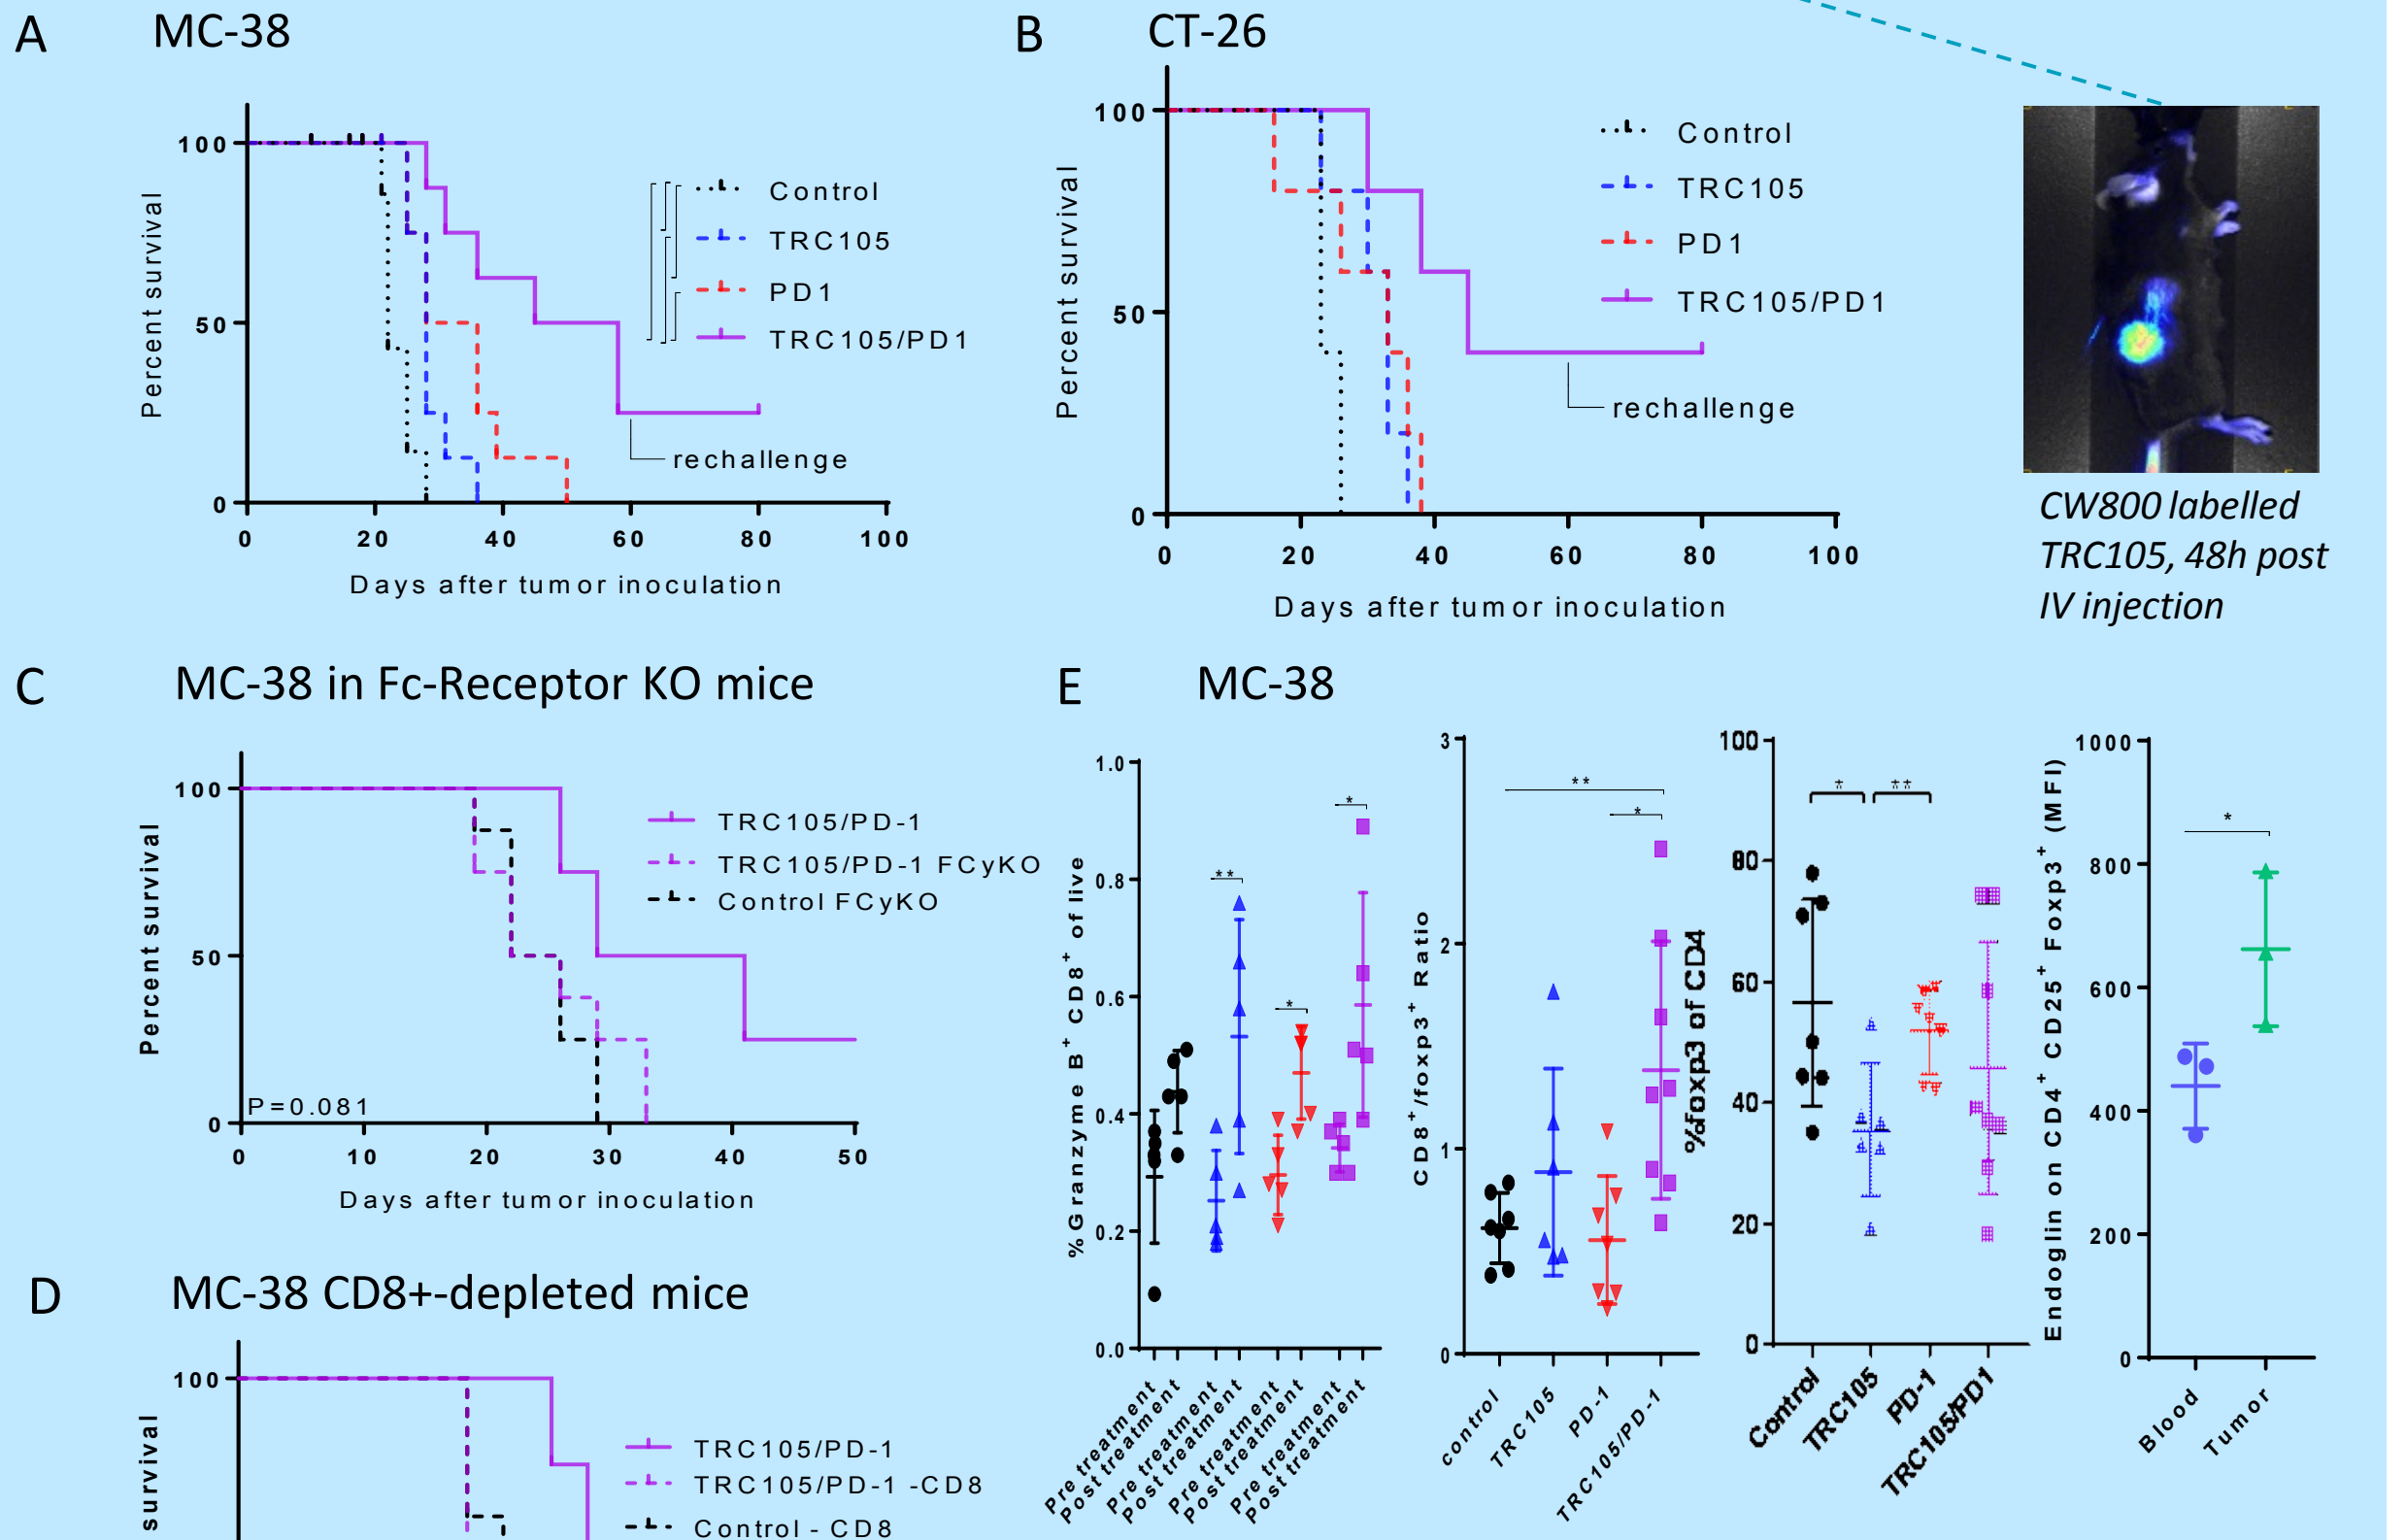
A MC-38
Percent survival
Control
TRC105
PD1
TRC105/PD1
rechallenge
Days after tumor inoculation
B CT-26
Percent survival
Control
TRC105
PD1
TRC105/PD1
rechallenge
Days after tumor inoculation
C MC-38 in Fc-Receptor KO mice
TRC105/PD-1
TRC105/PD-1 FCyKO
Control FCyKO
P=0.081
Days after tumor inoculation
D MC-38 CD8+-depleted mice
TRC105/PD-1
TRC105/PD-1 -CD8
Control - CD8
P=0.0017
Days after tumor inoculation
E MC-38
%Granzyme B+ CD8+ of live
CD8+/foxp3+ Ratio
%foxp3 of CD4
Endoglin on CD4+ CD25+ Foxp3+ (MFI)
Blood
Tumor

CW800 labelled TRC105, 48h post IV injection

Figure 3 Mice with subcutaneous MC38 (A) or CT26 (B) tumor treated with a combination of TRC105 and anti-PD1 therapy showed significantly increased survival and resistance to a rechallenge with tumor cells. These effects were dependent on the presence of Fc receptors (C), CD8+ T-cells (D) and involves an increase in activated CD8 cells together with a decrease in endoglin-expressing FoxP3+ Treg cells (E).

Conclusions:

- TRC105/PD1 significantly reduced number of lesions in a model of early stage CRC
- TRC105/PD1 combination therapy reduced tumor growth and increased survival
- Effects of TRC105/PD1 combination therapy was dependent on ADCC responses and the presence of CD8+ T cells
- TRC105 significantly reduced tumor resident endoglin- and FoxP3-expressing regulatory T-cells (Treg)

Contact: L.J.A.C.Hawinkels@LUMC.nl

This research was supported by a sponsored research grant from TRACON PHARMA