Biomarker Modulation in Patients Treated with TRC105 in Combination with anti-VEGF Therapy: Summary of Three Phase II Trials

Yingmiao Liu[1], Zhenhua Yuan[2], Dadong Zhang[2], Mark D. Starr[1], Chris Brady[1], Manoj Jivani[3], Bonne Adams[3], Delia Alvarez[3], Steven Attia[4], Toni Choueiri[5], Charles P. Theuer[3], Kouros Owzar[2], Herbert I. Hurwitz[1], Andrew B. Nixon[1]

[1]Duke University Medical Center, Durham, NC; [2]Duke Cancer Institute, Durham, NC; [3]Tracon Pharmaceuticals, Inc., San Diego, CA; [4]Mayo Clinic, Jacksonville, FL; [5]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA

Abstract #11546

Background

❖ TRC105 is an endoglin-targeting monoclonal antibody, with anti-angiogenic and anti-tumor activity[1]. Co-administration of TRC105 and bevacizumab (BEV) demonstrated activity in a phase Ib trial[2].

❖ Previously, we investigated biomarker expression at baseline and on-treatment related change in the phase Ib trial evaluating TRC105 and BEV[2]. TRC105 induced a dose-dependent increase in soluble endoglin levels. Unique biomarker features were observed in patients (pts) benefiting from this drug regimen.

❖ In this study, we expanded our biomarker analysis to 3 different phase II trials, where pts received TRC105 and different VEGF inhibitors to confirm previous observations. In addition to evaluating angiogenic and inflammatory markers, we assessed potential drug effects on multiple TGF-β family members.

Methods

❖ Plasma samples were collected from pts on three phase II trials combining TRC105 with an anti-VEGF agent:
•Axitinib in metastatic renal cell carcinoma (mRCC)[3]
•Pazopanib in advanced soft tissue sarcoma[4]
•Bevacizumab in glioblastoma (GBM)[5]

❖ Samples were processed at individual sites and shipped to the Duke Molecular Reference Laboratory for analysis.

❖ Samples were thawed one time, precipitate removed, aliquotted and re-frozen until assayed. All assays were performed in duplicate.

❖ Baseline and on-treatment change of 22 soluble protein biomarkers were assessed using Aushon BioSystems CiraScan protein multiplex arrays (Table 2).

Results

	mRCC	Sarcoma	GBM
n	18	18	22
Age, median(range)	62 (35-77)	58 (28-73)	59 (35-73)
Male, n (%)	16 (89%)	8 (44%)	16 (73%)
VEGF inhibitor dosing	Axitinib (5 mg/kg BID)	Pazopanib (800mg/day)	BEV (10mg/kg/2w)
VEGF inhibitor administration	C1D1	C1D1	C1D1
Lead in monotherapy	no	pazopanib lead-in for 1 month	BEV lead-in for 1 week
TRC105 dosing	8 or 10 mg/kg/w	8 or 10 mg/kg/w	8 or 10 mg/kg/w
TRC105 administration	C1D1	C2D1	C1D8
Response	5 PR (RECIST) 13 SD/PD	6 CR/PR (Choi) 12 SD/PD	0 PR or SD 22 PD

Table 1. Patient characteristics, drug dosing and drug schedules. Biomarker levels were assessed across 3 different clinical trials of TRC105. Patient characteristics, drug dosing/timing and overall response rates are shown. CR = complete response; PR = partial response; SD = stable disease; PD = progressive disease.

Biomarker	Unit	mRCC Median	mRCC Range	Sarcoma Median	Sarcoma Range	GBM Median	GBM Range
ANG2	pg/ml	161.2	3.0-305.4	123.9	12.9-502.4	75.7	6.9-142.1
BMP9	pg/ml	78.6	23.6-135.7	90.2	56.3-205.1	65.0	32.8-127.3
Endoglin	ng/ml	36.8	31.7-54.6	40.4	29.2-51.6	28.4	20.9-36.4
HGF	pg/ml	265.4	99.7-8156.5	368.5	38.2-11229.2	210.9	74.3-449.3
ICAM-1	ng/ml	584.8	405.4-923.2	657.9	292.8-1154.4	386.6	231.9-557.8
IL6	pg/ml	52.5	13.7-450.9	27.4	5.4-205.1	17.4	3.6-693.6
OPN	µg/ml	1.2	0.2-59.2	0.6	0.3-1.6	0.8	0.3-2.1
PDGFaa	pg/ml	422.1	19.6-5600.3	108.9	8.9-3006.1	216.5	40.4-3656.8
PDGFbb	pg/ml	847.7	25.4-3617.4	352.8	6.7-2941.3	943.3	142.4-3200.0
PIGF	pg/ml	22.8	13.5-198.0	16.5	4.5-77.3	30.9	14.3-43.6
SDF1	ng/ml	3.2	0.2-6.2	4.0	0.6-9.6	4.6	1.6-11.7
TGFβ1	ng/ml	42.2	10.0-205.7	25.4	10.7-296.6	36.4	13.6-228.1
TGFβ2	pg/ml	58.0	32.4-149.8	60.3	25.4-313.1	73.1	30.1-240.6
TGFβR3	ng/ml	182.0	115.5-233.7	133.5	70.7-271.2	173.1	87.6-268.9
TIMP	ng/ml	119.3	53.0-382.3	88.8	48-213.7	56.8	36.6-110.1
TSP2	ng/ml	124.9	48.4-475.9	82.4	58.3-281.2	68.0	40.7-140.5
VCAM1	µg/ml	1.7	1.0-3.1	1.5	0.6-2.8	2.0	1.4-3.5
VEGF	pg/ml	185.4	49.3-885.3	51.9	1.8-393.3	114.2	37.9-236.0
VEGFD	ng/ml	1.1	0.6-1.8	1.0	0.6-11.5	0.9	0.6-2.3
VEGFR1	pg/ml	64.4	28.3-660.5	318.0	25.5-1343.8	49.7	31.5-244.5
VEGFR2	ng/ml	4.1	1.6-5.2	4.9	2.6-16.8	3.8	2.3-6.2
VEGFR3	ng/ml	377.3	162.8-647.3	306.7	99-553.6	176.8	88.1-348.0

Table 2. Biomarker baseline levels. Median and range for all biomarkers at baseline are shown.

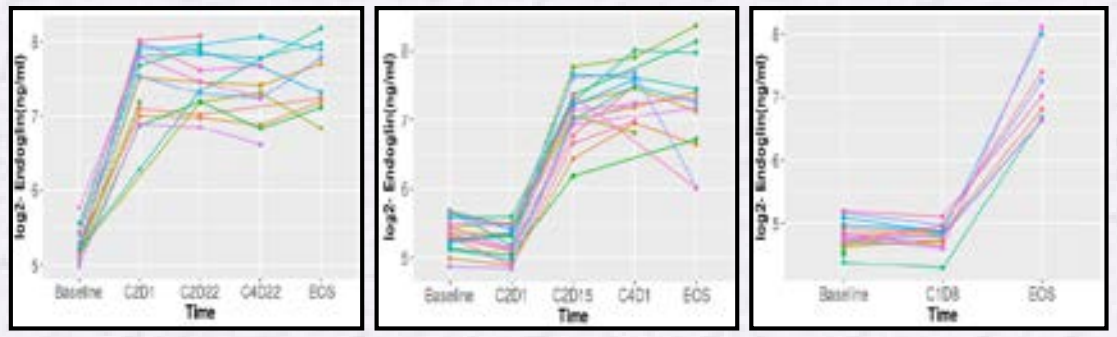
mRCC
Sarcoma
GBM
log2- Endoglin(ng/ml)
Baseline
C2D1
C2D22
C4D22
EOS
C2D15
C4D1
C1D8
Time

Figure 1. Change in endoglin levels from baseline through EOS. Each line represents an individual patient. Increases in soluble endoglin is dependent upon treatment with TRC105. Significant increases of endoglin were observed in all 3 patient populations after TRC105 administration. Differences in the timing of endoglin increases are due to the different dosing schedules of TRC105 across the 3 trials (see Table 1).

Response Analysis

❖ Drug activity was observed in two of the trials (Table 1).
- In mRCC, 5 pts exhibited partial response (RECIST1.1 criteria) after treatment with the combination of TRC105 and axitinib.
- In sarcoma, 6 pts exhibited responses by Choi criteria after treatment with the combination of TRC105 and pazopanib.

❖ In studies where patients exhibited responses, potential biomarkers differentiating responders versus non-responders were explored. Biomarkers associated with response were identified at both baseline and on-treatment (Figure 3).

Biomarker	mRCC (C2D1) Effect size	mRCC (C2D1) p-value	Sarcoma (C2D15) Effect size	Sarcoma (C2D15) p-value	GBM (C2D15) Effect size	GBM (C2D15) p-value
ANG2	-1.77	<0.001	-1.51	<0.001	-1.01	0.06
BMP9	-0.10	0.45	-0.16	0.04	0.13	0.44
Endoglin	2.18	<0.001	1.77	<0.001	2.23	0.03
HGF	-0.17	0.06	0.51	0.02	0.06	0.69
ICAM1	-0.11	0.11	-0.13	0.02	0.10	0.31
IL6	0.003	0.98	0.50	0.04	0.37	0.69
OPN	-0.20	0.11	0.28	0.002	0.68	0.03
PDGFaa	0.37	0.72	0.40	0.39	-0.02	1.00
PDGFbb	0.33	0.64	0.68	0.13	0.19	0.69
PIGF	1.50	<0.001	1.37	<0.001	-0.12	0.06
SDF1	0.36	0.003	0.19	0.35	0.13	0.69
TGFβ1	0.25	0.45	0.55	0.25	-0.05	1.00
TGFβ2	0.004	0.93	0.20	0.13	-0.07	1.00
TGFβR3	-0.25	0.03	-0.31	0.01	0.17	0.31
TIMP1	-0.15	0.30	-0.14	0.20	0.34	0.31
TSP2	-0.37	<0.001	-0.03	0.67	0.14	0.31
VCAM1	0.75	<0.001	0.64	<0.001	0.55	0.03
VEGF	1.65	<0.001	2.23	<0.001	0.07	0.84
VEGFD	0.20	0.003	0.21	0.04	0.03	0.69
VEGFR1	-0.20	0.23	0.04	0.87	0.31	0.09
VEGFR2	-0.62	<0.001	-0.38	<0.001	-0.03	1.00
VEGFR3	-1.03	<0.001	-0.80	<0.001	-0.20	0.06

Table 3. Modulation of biomarkers in response to TRC105 and VEGF inhibitors. Wilcoxon signed rank analyses are shown above. Significant biomarker modulation was observed in response to co-administration of both drugs. Time points analyzed for each study were:
- mRCC: Baseline to C2D1
- Sarcoma: Baseline to C2D15
- GBM: Baseline to C2D15

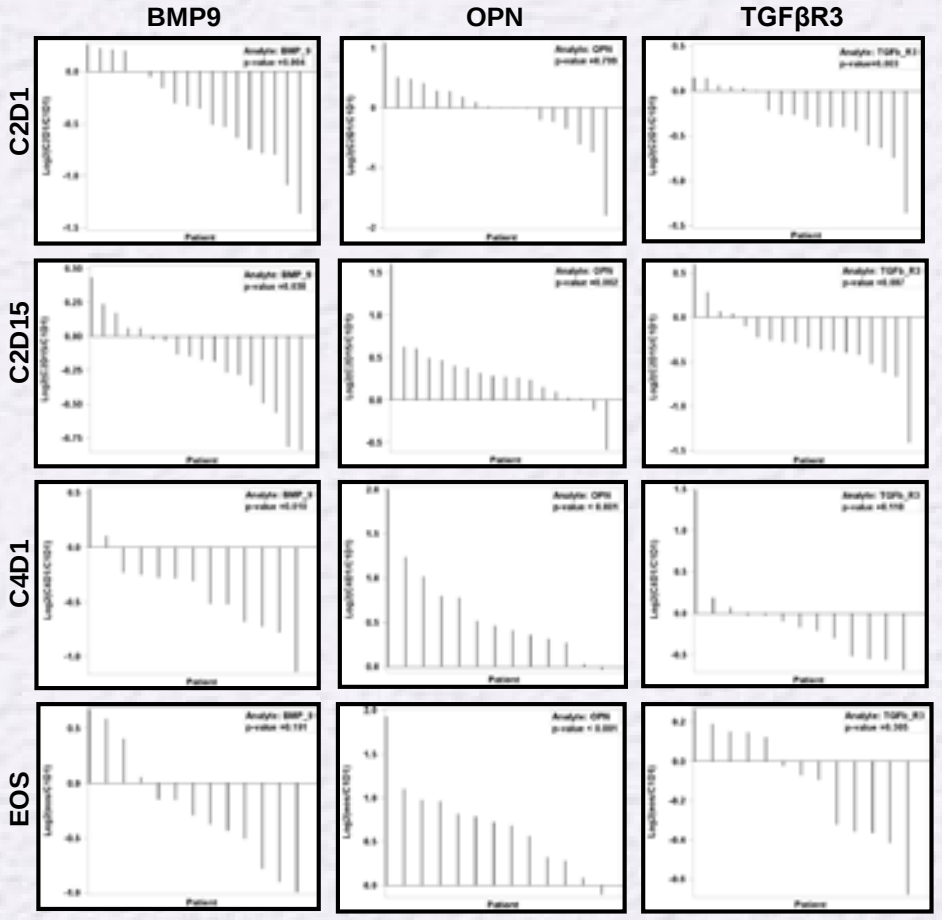
BMP9
OPN
TGFβR3
C2D1
C2D15
C4D1
EOS
Patient

Figure 2. Change in TGFβ family members over time after treatment in sarcoma pts. TRC105 targets endoglin, a type III receptor for TGFβ. We included additional TGFβ family members in our biomarker analysis. Significant changes in BMP9, TGFβR3, and OPN were observed in sarcoma pts treated with TRC105 and pazopanib.

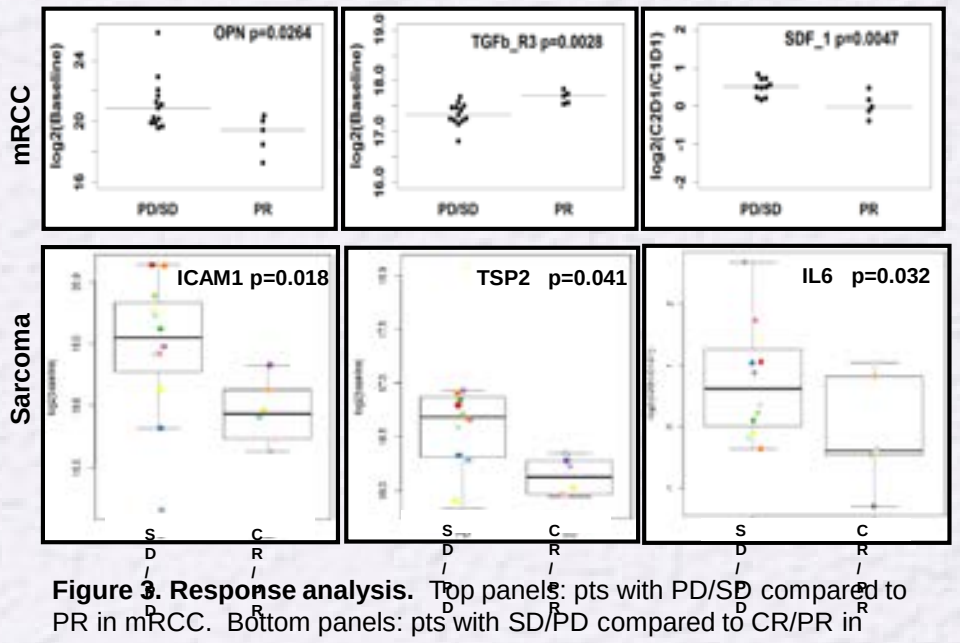
Baseline
Baseline
C2D1
mRCC
Sarcoma
OPN p=0.0264
TGFb_R3 p=0.0028
SDF_1 p=0.0047
PD/SD
PR
ICAM1 p=0.018
TSP2 p=0.041
IL6 p=0.032

Figure 3. Response analysis. Top panels: pts with PD/SD compared to PR in mRCC. Bottom panels: pts with SD/PD compared to CR/PR in sarcoma. The left four panels reflect differences at baseline; the right two panels reflect the change from baseline to C2D1. A dichotomized rank sum test was used to identify differences in biomarker expression at baseline as well as on-treatment change between responders and non-responders.

Conclusions

❖ This report is the first analysis across multiple tumor types evaluating circulating biomarkers from cancer pts treated with TRC105 in combination with a VEGF inhibitor.

- Baseline levels and on-treatment change for most markers were similar to those observed in the original phase Ib TRC105 + BEV trial.
- Baseline variation was observed for PDGFaa, PDGFbb, VEGF and VEGFR1, possibly reflecting differences in the underlying tumor type or patient population.

❖ Soluble endoglin consistently increases after TRC105 administration, confirming its role as a pharmacodynamic marker of TRC105 treatment.

❖ Baseline and on-treatment change analyses identified markers differentiating responders from non-responders. Lower baseline expression of OPN and higher baseline expression of TGFβR3 were associate with better outcomes in mRCC. Lower baseline expression of ICAM-1 and TSP-2 in sarcoma pts were associated with better outcomes.

❖ Significant changes in TGF-β family members were observed in sarcoma pts, but not in mRCC or GBM pts. The potential prognostic and predictive effects of these markers in sarcoma will be confirmed in the ongoing phase III trial of TRC105 and pazopanib in angiosarcoma (NCT02979899).

References

1.Fonsatti et al, 2010, Cardiovascular Res, 86(1): 12-19
2.Liu et al, 2014, J Clin Oncol 32:5s, 2014 (suppl; abstr 11020)
3.Choueiri et al, 2015, 14th International Kidney Cancer Symposium
4.Attia et al, 2015, J Clin Oncol 33, 2015 (suppl; abstr 10514)
5.Ahluwalia et al, 2016, J Clin Oncol 34, 2016 (suppl; abstr 2035)

Acknowledgements

We gratefully acknowledge the invaluable contributions of the patients, their families, and the research staff who participated in these studies. Research funding was provided by Tracon Pharmaceuticals.